PS-1001: A NOVEL PAN-INFLAMMASOME INHIBITOR TARGETING ASC FOR THE TREATMENT OF HIDRADENITIS SUPPURATIVA

Nisha Padmanabhan[1], Edwin K Lim[1], Abigail YT Loh[1], Qihui Seet[1], Amir M Hossini[2], Christos C Zouboulis[2], Mark Cockett[3], Lewis Z Hong[1]

[1]Paratus Sciences, Singapore, [2]Departments of Dermatology, Venereology, Allergology, and Immunology, Staedtisches Klinikum Dessau, Brandenburg Medical School Theodor Fontane and Faculty of Health Sciences Brandenburg, Dessau, Germany, [3]Paratus Sciences, New York, USA

Background & Rationale

Therapeutic Rationale in HS

High levels of DAMPs/PAMPs in HS lesions drive chronic inflammasome activation. By targeting ASC, PS-1001 delivers pan-inflammasome blockade, preventing the release of both IL-1β and IL-18 and arresting the inflammatory cascade at its source.

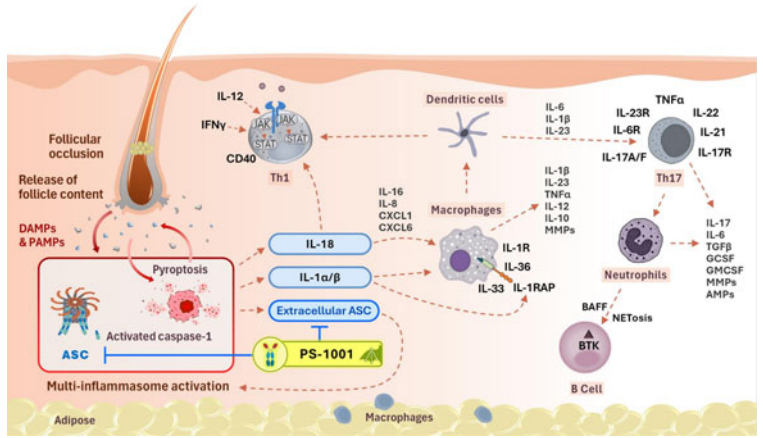
Follicular occlusion
Release of follicle content
DAMPs & PAMPs
Pyroptosis
Activated caspase-1
ASC
PS-1001
Multi-inflammasome activation
Adipose
Macrophages
IL-12
IFNγ
CD40
Th1
Dendritic cells
IL-18
IL-1α/β
Extracellular ASC
Macrophages
IL-16
IL-8
CXCL1
CXCL6
IL-1R
IL-36
IL-33
IL-1RAP
TNFα
IL-23R
IL-6R
IL-17A/F
Th17
IL-22
IL-21
IL-17R
Neutrophils
NETosis
BAFF
BTK
B Cell

PS-1001 Molecule Profile

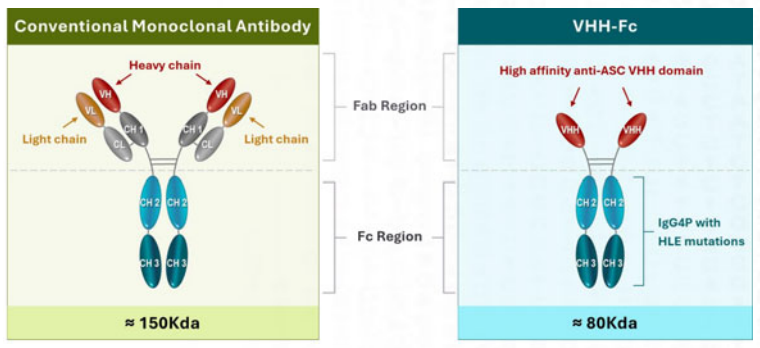
Conventional Monoclonal Antibody
Heavy chain
Light chain
Light chain
Fab Region
Fc Region
≈ 150Kda
VHH-Fc
High affinity anti-ASC VHH domain
IgG4P with HLE mutations
≈ 80Kda

- **Structure:** PS-1001 is a bivalent VHH-Fc fusion protein (80 kDa) consisting of a high affinity anti-ASC VHH domain fused to a human IgG4P Fc backbone engineered with half-life extending mutations.
- **Tissue Penetration:** PS-1001's smaller format (compared to mAbs) facilitates deep tissue penetration into fibrotic, indurated HS lesions.
- **Intracellular Delivery:** PS-1001 was engineered for intracellular uptake and cellular potency, ensuring potent target engagement.

Dual Mechanism of Action

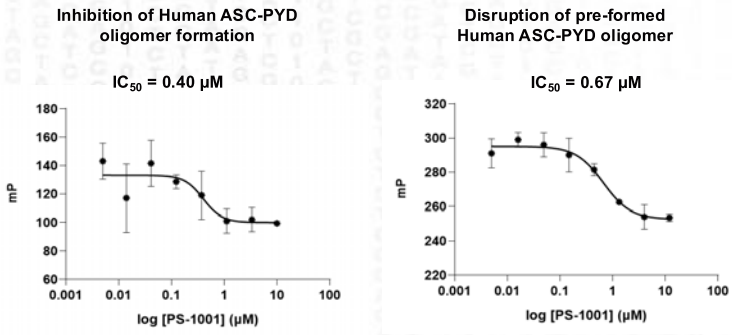
Inhibition of Human ASC-PYD oligomer formation
IC50 = 0.40 μM
mP
log [PS-1001] (μM)
Disruption of pre-formed Human ASC-PYD oligomer
IC50 = 0.67 μM
mP
log [PS-1001] (μM)

PS-1001 is a pan-inflammasome inhibitor that targets ASC via two distinct modes of action:

- Inhibition of Speck Formation: Inhibits nucleation of productive ASC filament formation, preventing inflammasome activation, pyroptosis, and cytokine release (IL-1β and IL-18).
- Disruption of Pre-formed Specks: Actively disassembles existing ASC filaments—both intracellular and extracellular—dampening the spread of inflammation.

Pharmacologic Profile and In Vivo Proof-of-Concept

Potent Cellular Activity

PS-1001 potently inhibits IL-1β and IL-18 release in inflammasome-activated (LPS:ATP) human PBMCs.

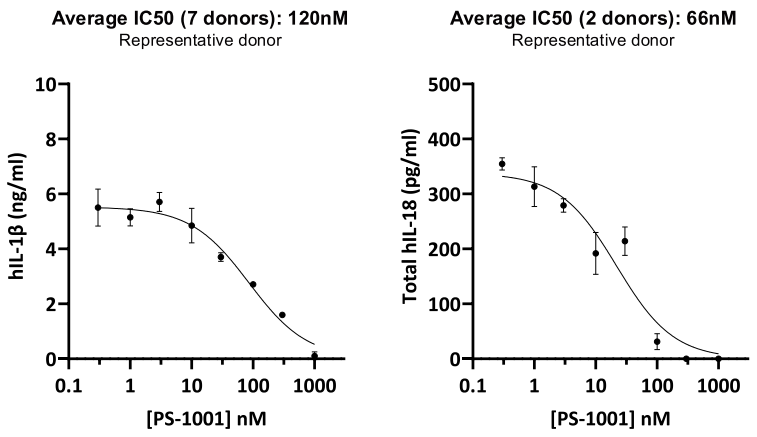
Average IC50 (7 donors): 120nM
Representative donor
hIL-1β (ng/ml)
[PS-1001] nM
Average IC50 (2 donors): 66nM
Representative donor
Total hIL-18 (pg/ml)
[PS-1001] nM

Biodistribution and Skin Bioavailability

Following systemic administration (30mg/kg IV) in C57BL/6 wild type mice, PRT-000112* exhibits extensive tissue distribution, achieving high concentrations in the skin, the primary target tissue for HS.

*PRTa-000112 retains the same VHH sequence of PS-1001 without half-life extended mutations in Fc backbone

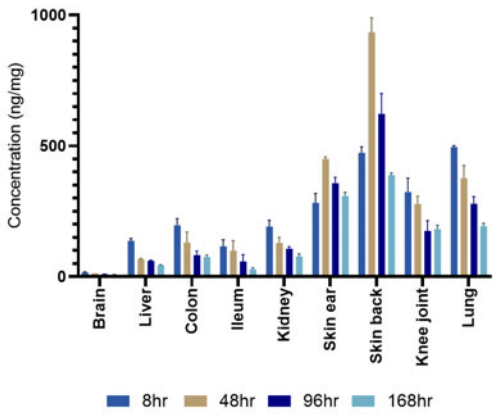
Concentration (ng/mg)
Brain
Liver
Colon
Ileum
Kidney
Skin ear
Skin back
Knee joint
Lung
8hr
48hr
96hr
168hr

Efficacy in Inflammatory Models

In Vivo Efficacy. Efficacy in Skin Inflammation Model: PS-1001 significantly reduced disease pathology and inflammation in the Imiquimod (IMQ)-induced psoriasis model.

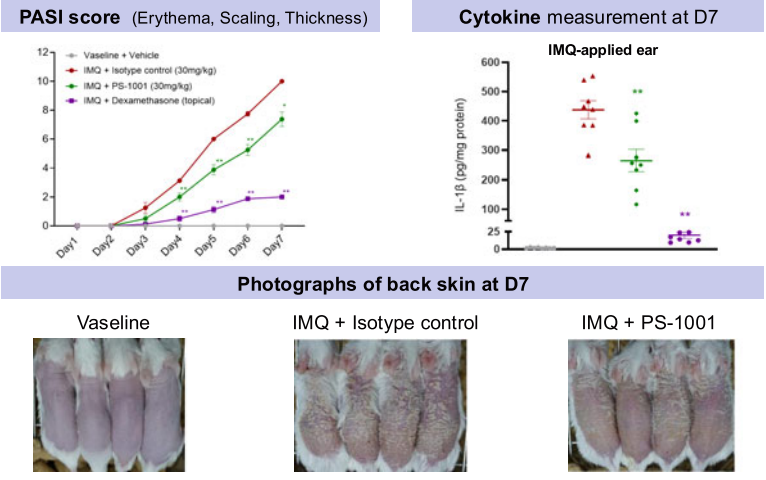
PASI score (Erythema, Scaling, Thickness)
Vaseline + Vehicle
IMQ + Isotype control (30mg/kg)
IMQ + PS-1001 (30mg/kg)
IMQ + Dexamethasone (topical)
Cytokine measurement at D7
IMQ-applied ear
IL-1β (pg/mg protein)
Photographs of back skin at D7
Vaseline
IMQ + Isotype control
IMQ + PS-1001

- **Broad Activity:** Efficacy was also observed in Gout (reduced joint swelling/pain) and Colitis models (reduced DAI score), confirming robust anti-inflammatory activity in different disease contexts.

Efficacy in Patient-Derived HS 3D-SeboSkin Model

Preclinical Proof-of-Concept in Human Tissue: PS-1001 was evaluated in the HS 3D-SeboSkin model, an *ex vivo* system derived directly from lesional skin of HS patients. This model preserves the native immune architecture and inflammatory milieu of the disease.

Potent IL-18 Inhibition

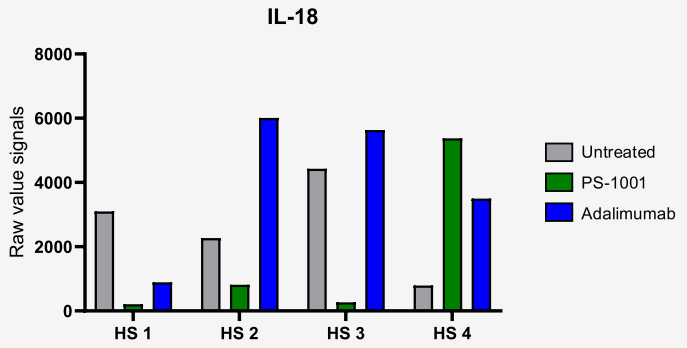
IL-18
Raw value signals
HS 1
HS 2
HS 3
HS 4
Untreated
PS-1001
Adalimumab

- **Robust Response:** PS-1001 achieved a >60% reduction in IL-18 secretion compared to vehicle in 3/4 patient donors.
- **Target Engagement:** Significant suppression of IL-18 confirms PS-1001 effectively penetrates HS tissue and inhibits inflammasome activity in HS skin.
- **Distinct Mechanism:** IL-18 is a key inflammatory driver in HS that is not addressed by IL-1 inhibitors or adalimumab (TNF-α blockade).

Broader Inflammatory Activity than Adalimumab

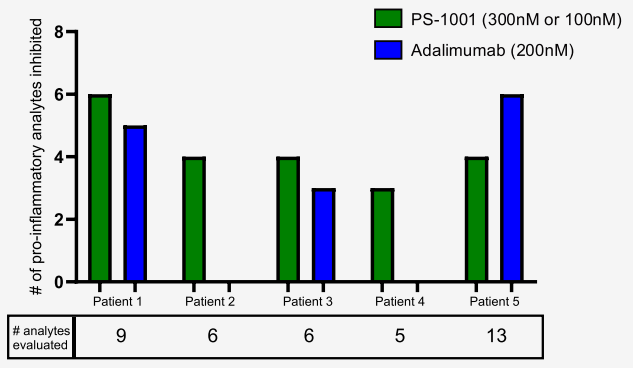
PS-1001 (300nM or 100nM)
Adalimumab (200nM)
of pro-inflammatory analytes inhibited
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5
analytes evaluated
9
6
6
5
13

- **Superior Breadth:** PS-1001 suppressed a significantly broader repertoire of pro-inflammatory analytes compared to the standard of care, adalimumab.
- **Upstream vs Downstream:** While adalimumab targets a single downstream cytokine (TNF-α), PS-1001 inhibits the inflammasome upstream, abrogating the release of multiple DAMP/PAMP-driven cytokines simultaneously.

Dampening of Distinct Innate and Adaptive Pathogenic Axes

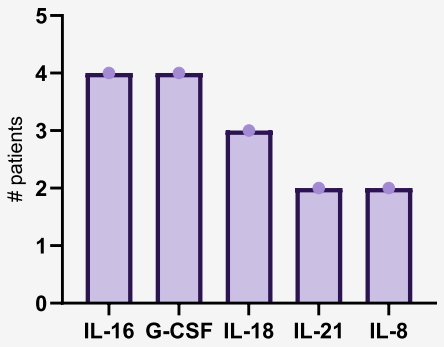
patients
IL-16
G-CSF
IL-18
IL-21
IL-8

- **Neutrophil Axis (IL-8, G-CSF):** Suppressing these chemoattractants limits neutrophil infiltration, a key driver of abscess formation and purulence.
- **Chronicity Signals (IL-21, IL-16):** Inhibition of these cytokines limits T-cell recruitment and maintenance, potentially disrupting the Th17 axis associated with disease persistence.
- **Th1/IFN-γ Axis (IL-18):** Reduction of IL-18 disrupts a critical driver of IFN-γ production and the Th1 inflammatory loop.

Safety, PK & Clinical Readiness

- **Non-Clinical Safety:** No adverse findings observed during a single-dose (30 mg/kg) NHP PK study over the 28-day observation period.
- **CMC and Developability:** High-concentration (>100 mg/mL) liquid formulation exhibits low viscosity and excellent stability, supporting a low-volume subcutaneous (SC) autoinjector presentation.
- **Immunogenicity Risk:** Favorable profile observed in *in vitro* assays (50 donors), demonstrating low T-cell activation potential comparable to approved biologics.

Predicted Human PK Supports Q4W Dosing Regimen

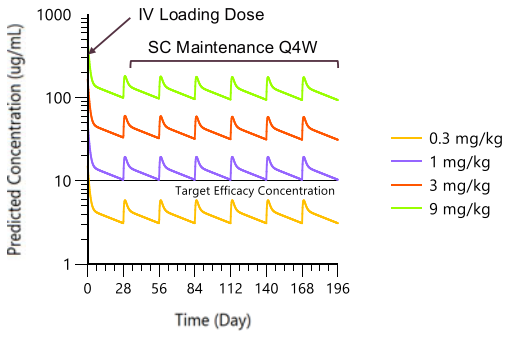
IV Loading Dose
SC Maintenance Q4W
Predicted Concentration (ug/mL)
Target Efficacy Concentration
Time (Day)
0.3 mg/kg
1 mg/kg
3 mg/kg
9 mg/kg

- **Modeling Strategy:** Human PK profiles were projected based on NHP PK parameters.
- **Half-Life Extension:** Engineered Fc backbone confers a predicted human half-life ($t_{1/2}$) of ~1180 hours (~49 days), validating the molecular design for chronic dosing.
- **Regimen Optimization:** Modeling confirms that an IV loading dose followed by SC maintenance supports a Q4W dosing interval, maintaining target trough coverage (C_{trough}) projected to be efficacious.

Conclusion

- **Novel Mechanism of Action:** PS-1001 validates ASC as a tractable therapeutic target, delivering potent pan-inflammasome inhibition.
- **Ex Vivo Proof of Mechanism in HS:** In patient-derived HS tissue, PS-1001 demonstrated a broader anti-inflammatory profile compared to adalimumab. By blocking ASC, the central component of the inflammasome complex, PS-1001 inhibits both IL-1β and IL-18, distinguishing it from selective IL-1 blockers that leave IL-18 unaddressed.
- **Candidate Profile:** Favorable PK and developability characteristics support a patient-friendly Q4W subcutaneous regimen.
- **Path to Clinic:** IND-enabling studies are currently underway to support the initiation of First-in-Human (FIH) clinical trials in late 2026.

Paratus SCIENCES